Gender Differences in Chronic Kidney Disease (CKD) Progression: Real-World Data (RWD) Analysis

Joseph Stavas[a], Guido Filler[b], Debra Irwin[c], Kate Lovett[c], Rohan Shah[c], Emily Butler[a], Libbie McKenzie[a], Maria Ferris[d]

ProKidney Raleigh NC, USA[a], London Health Science Centre London ON, CAN[b], Aetion New York NY, USA[c], University of North Carolina Chapel Hill NC, USA[d]

1. Introduction/Background

- The prevalence of CKD has been reported consistently as higher in women compared with men, and this difference has been consistent over time (15.4% v. 12.8% in 2011–2012). However, the incidence of end-stage kidney disease (ESKD) was 1.5 times higher and mortality was higher in men.[1,2]
- Studies used to make these claims have been limited by smaller numbers and possible selection bias through tertiary care centers[1].
- Large population-based studies utilizing RWD to evaluate gender related trends are scant
- If true, additional investigations are urgently needed to identify biologic and psychosocial factors responsible for the gender differences.

2. Goal

The goal of this analysis was to use Real-world Data (RWD) to evaluate gender difference in the initiation of dialysis.

3. Methods

- HealthVerity Private Source20 closed claims linked with Veradigm Health Insights EHR and Quest laboratory results data of adults compared progression of CKD stage ≥3b to dialysis, by gender.
- Patients were required to have ≥2 eGFR measurements 90-365 days apart and were followed until they initiated dialysis or end of available data. The first eGFR as the index date (baseline).
- We excluded pregnancy, acute kidney injury (AKI), ESKD and dialysis at baseline.
- Covariates included gender, country region, age at index date, Deyo- Chronic Comorbidity Index (CCI) Score, eGFRs, payer types and comorbidities.
- Please see attrition figure (Figure 1). Those in final study population in stages 3b-5 =14,072

Figure 1: Attrition Waterfall

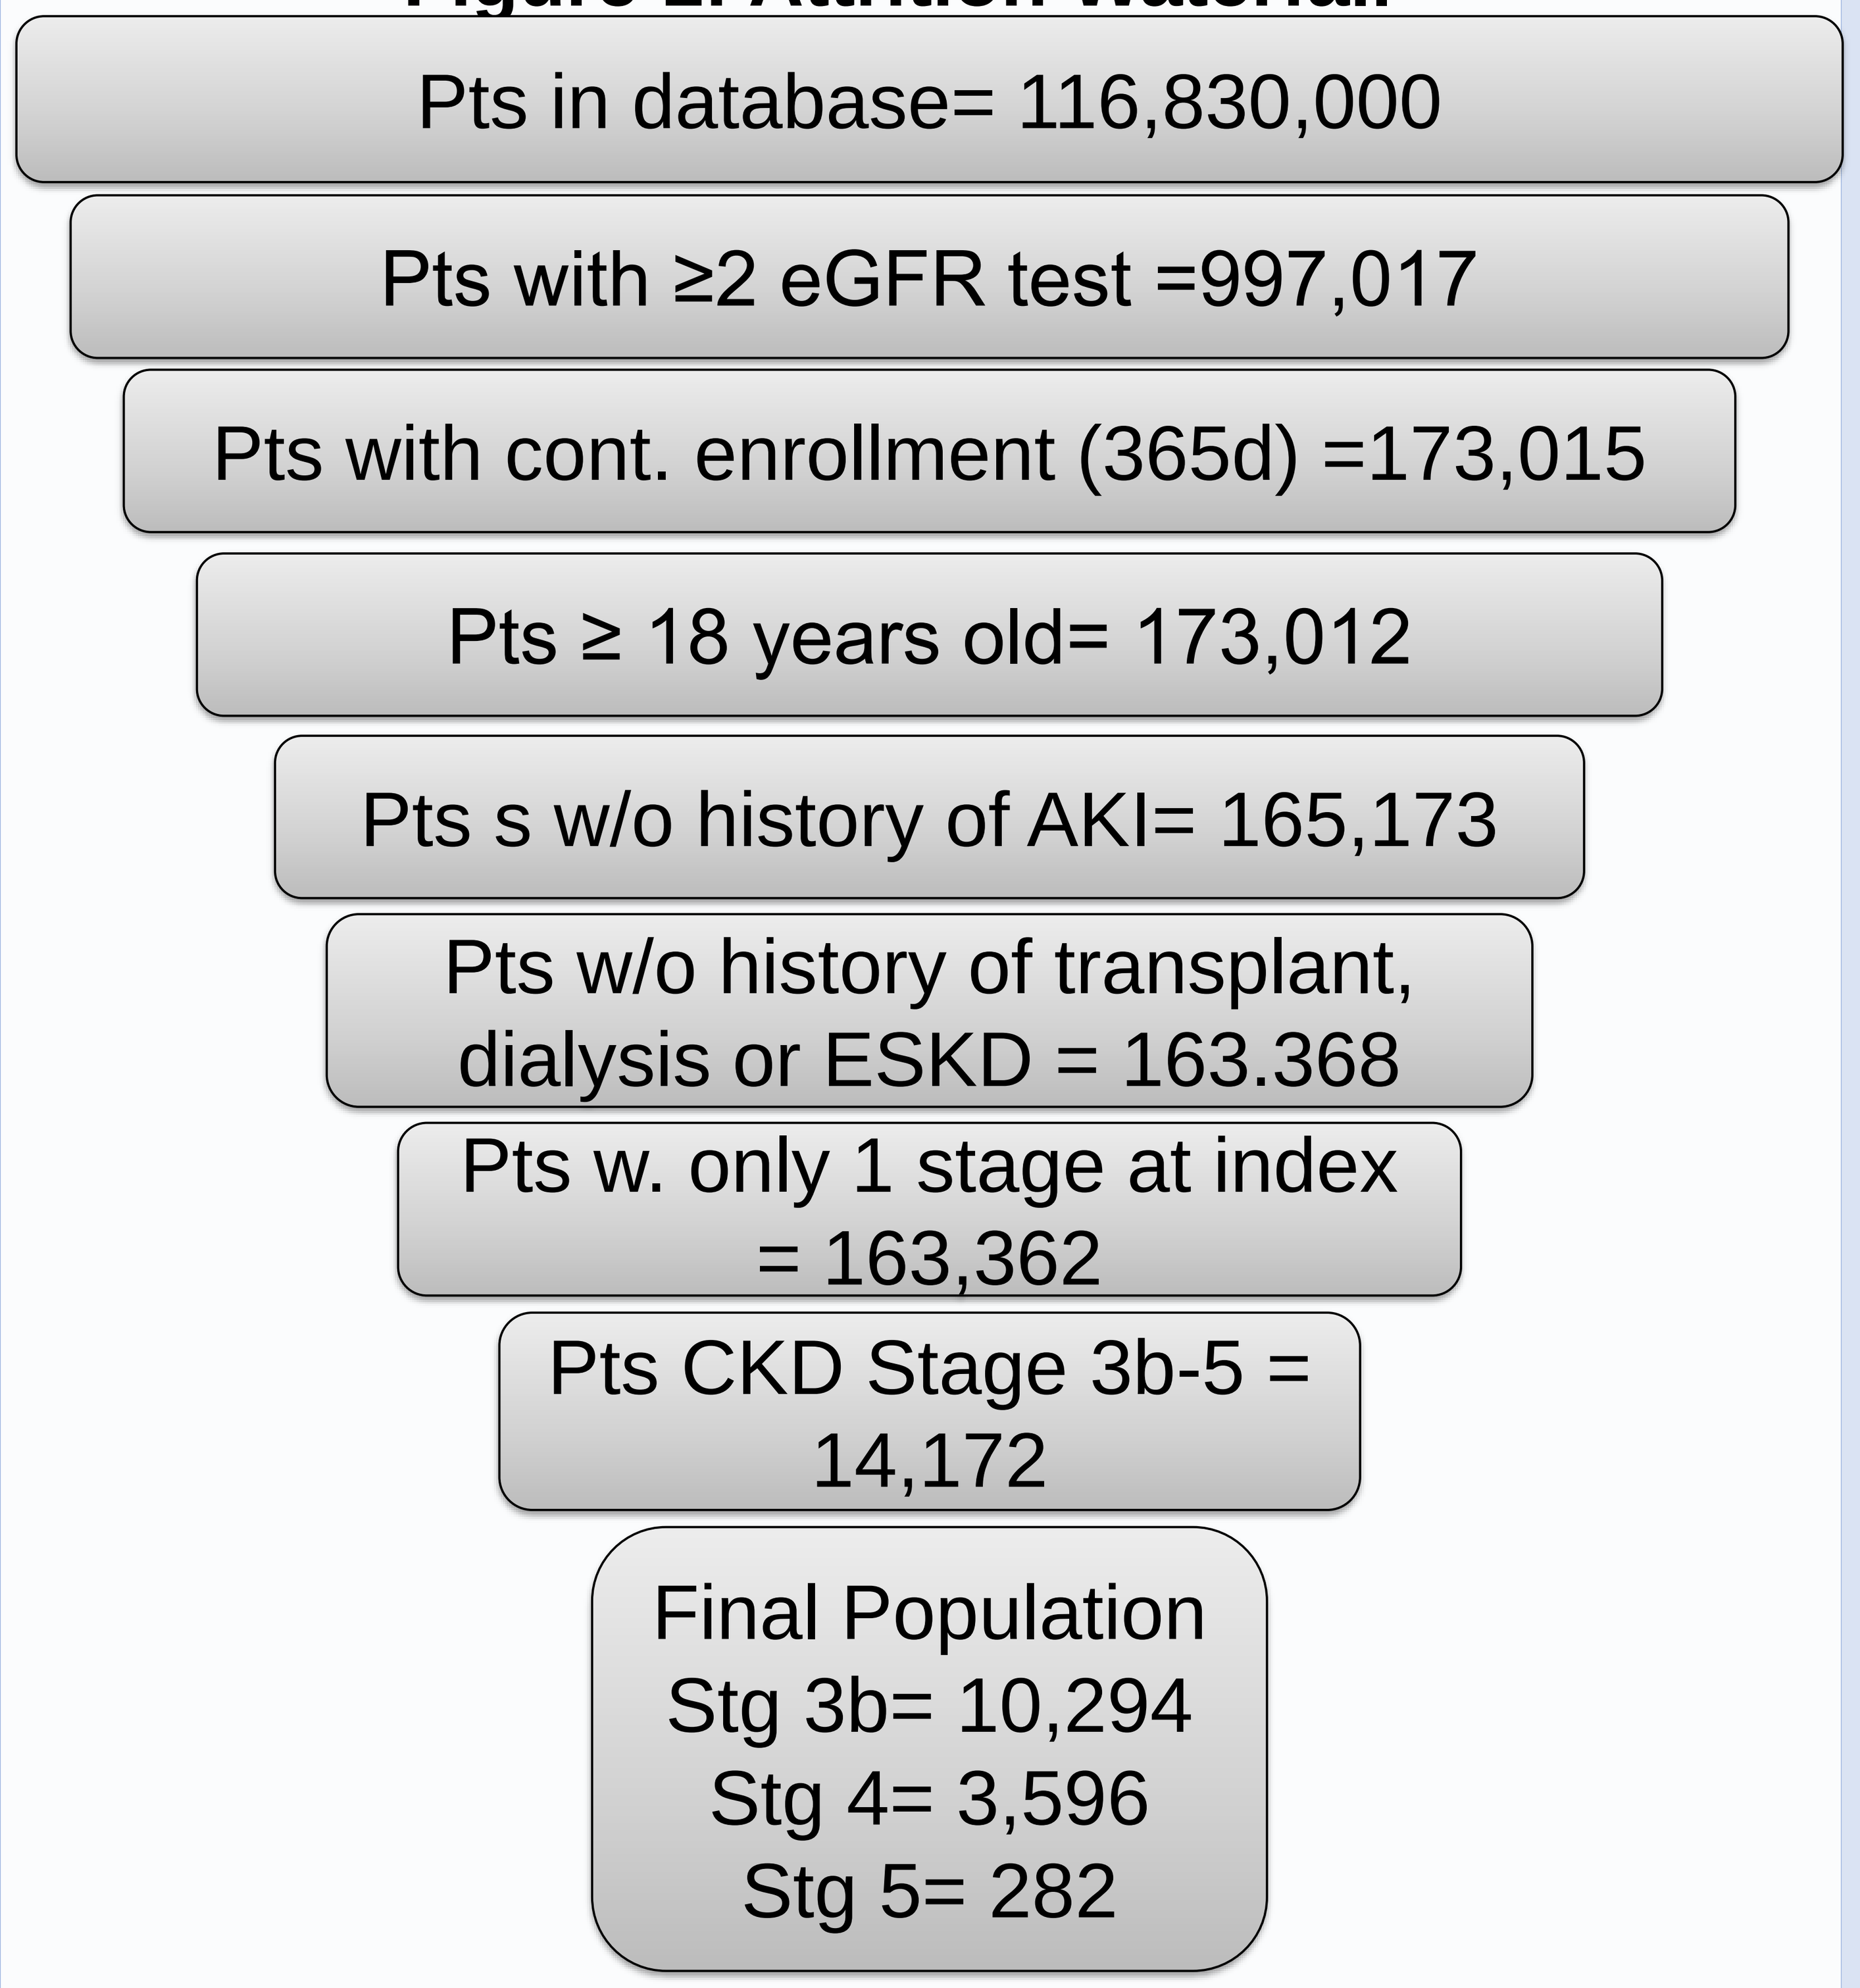
Pts in database= 116,830,000
Pts with ≥2 eGFR test =997,017
Pts with cont. enrollment (365d) =173,015
Pts ≥ 18 years old= 173,012
Pts s w/o history of AKI= 165,173
Pts w/o history of transplant, dialysis or ESKD = 163.368
Pts w. only 1 stage at index = 163,362
Pts CKD Stage 3b-5 = 14,172
Final Population
Stg 3b= 10,294
Stg 4= 3,596
Stg 5= 282

4. Results

- The mean age between men and women with CKD stage 3b-5 was similar (70 [61,78] v. 71 [63,79]).
- A significantly larger proportion of the study population (n=14,172) were women (56.3%) and this was consistent across all CKD stages(p<0.04).
- Mean eGFR results at entry were similar for men (34.1±8.0) and women(33.8±8.1).
- The type of health insurance differed between genders (p<0.05) With men more likely to have commercial (31.2% v 24/6%) and women more likely to have Medicaid (57.1 v. 63.0) .
- Men had higher Mean Deyo-CCI scores compared to women (p<0.05) and a higher proportion of renal osteodystrophy (2.2% vs 1.6%, p,0.05); while women were more likely to have anemia (21% vs 19.6%,p<0.05).
- For CKD stages ≥3b, the proportion of men who initiated dialysis was significantly higher (2.5% vs1.9%, p<0.05), and the mean time to initiation of dialysis was significantly shorter (510±340 vs 530±356 days, p<0.05) compared to women.

Figure 2: Percent of Patients who Initiated Dialysis

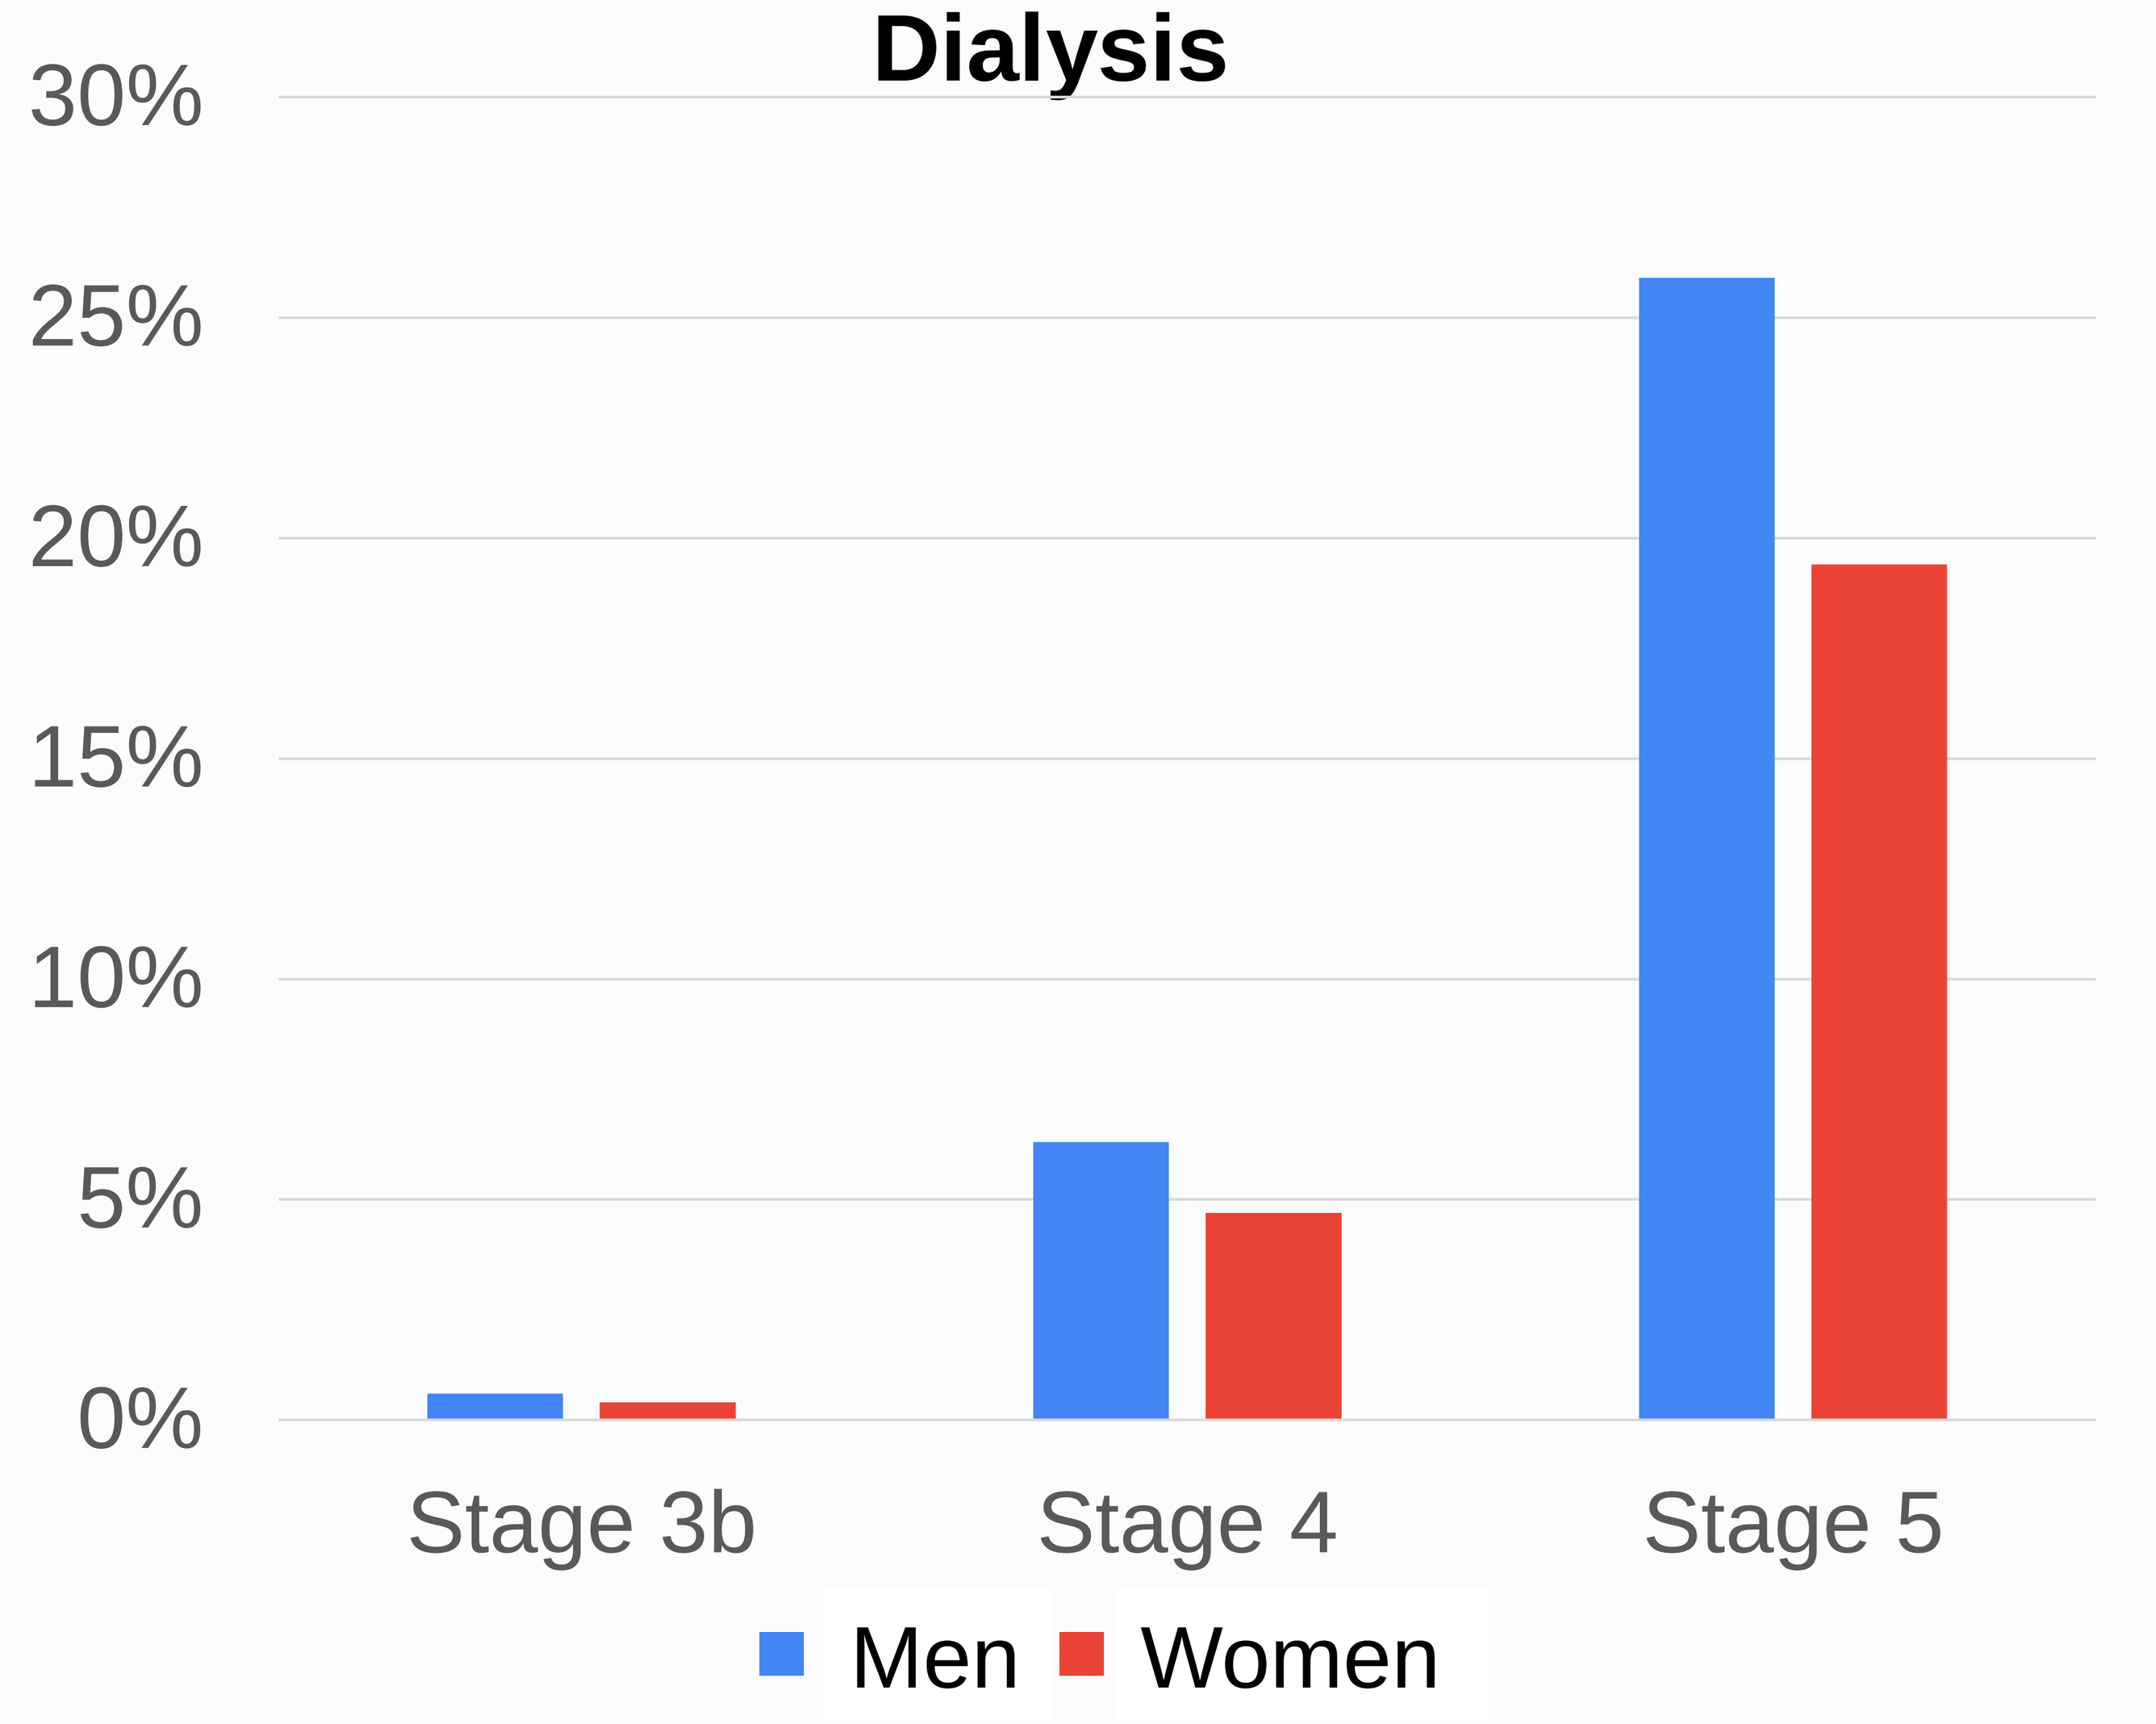
30%
25%
20%
15%
10%
5%
0%
Stage 3b
Stage 4
Stage 5
Men
Women

Figure 3: Time to Initiation of Dialysis in Days

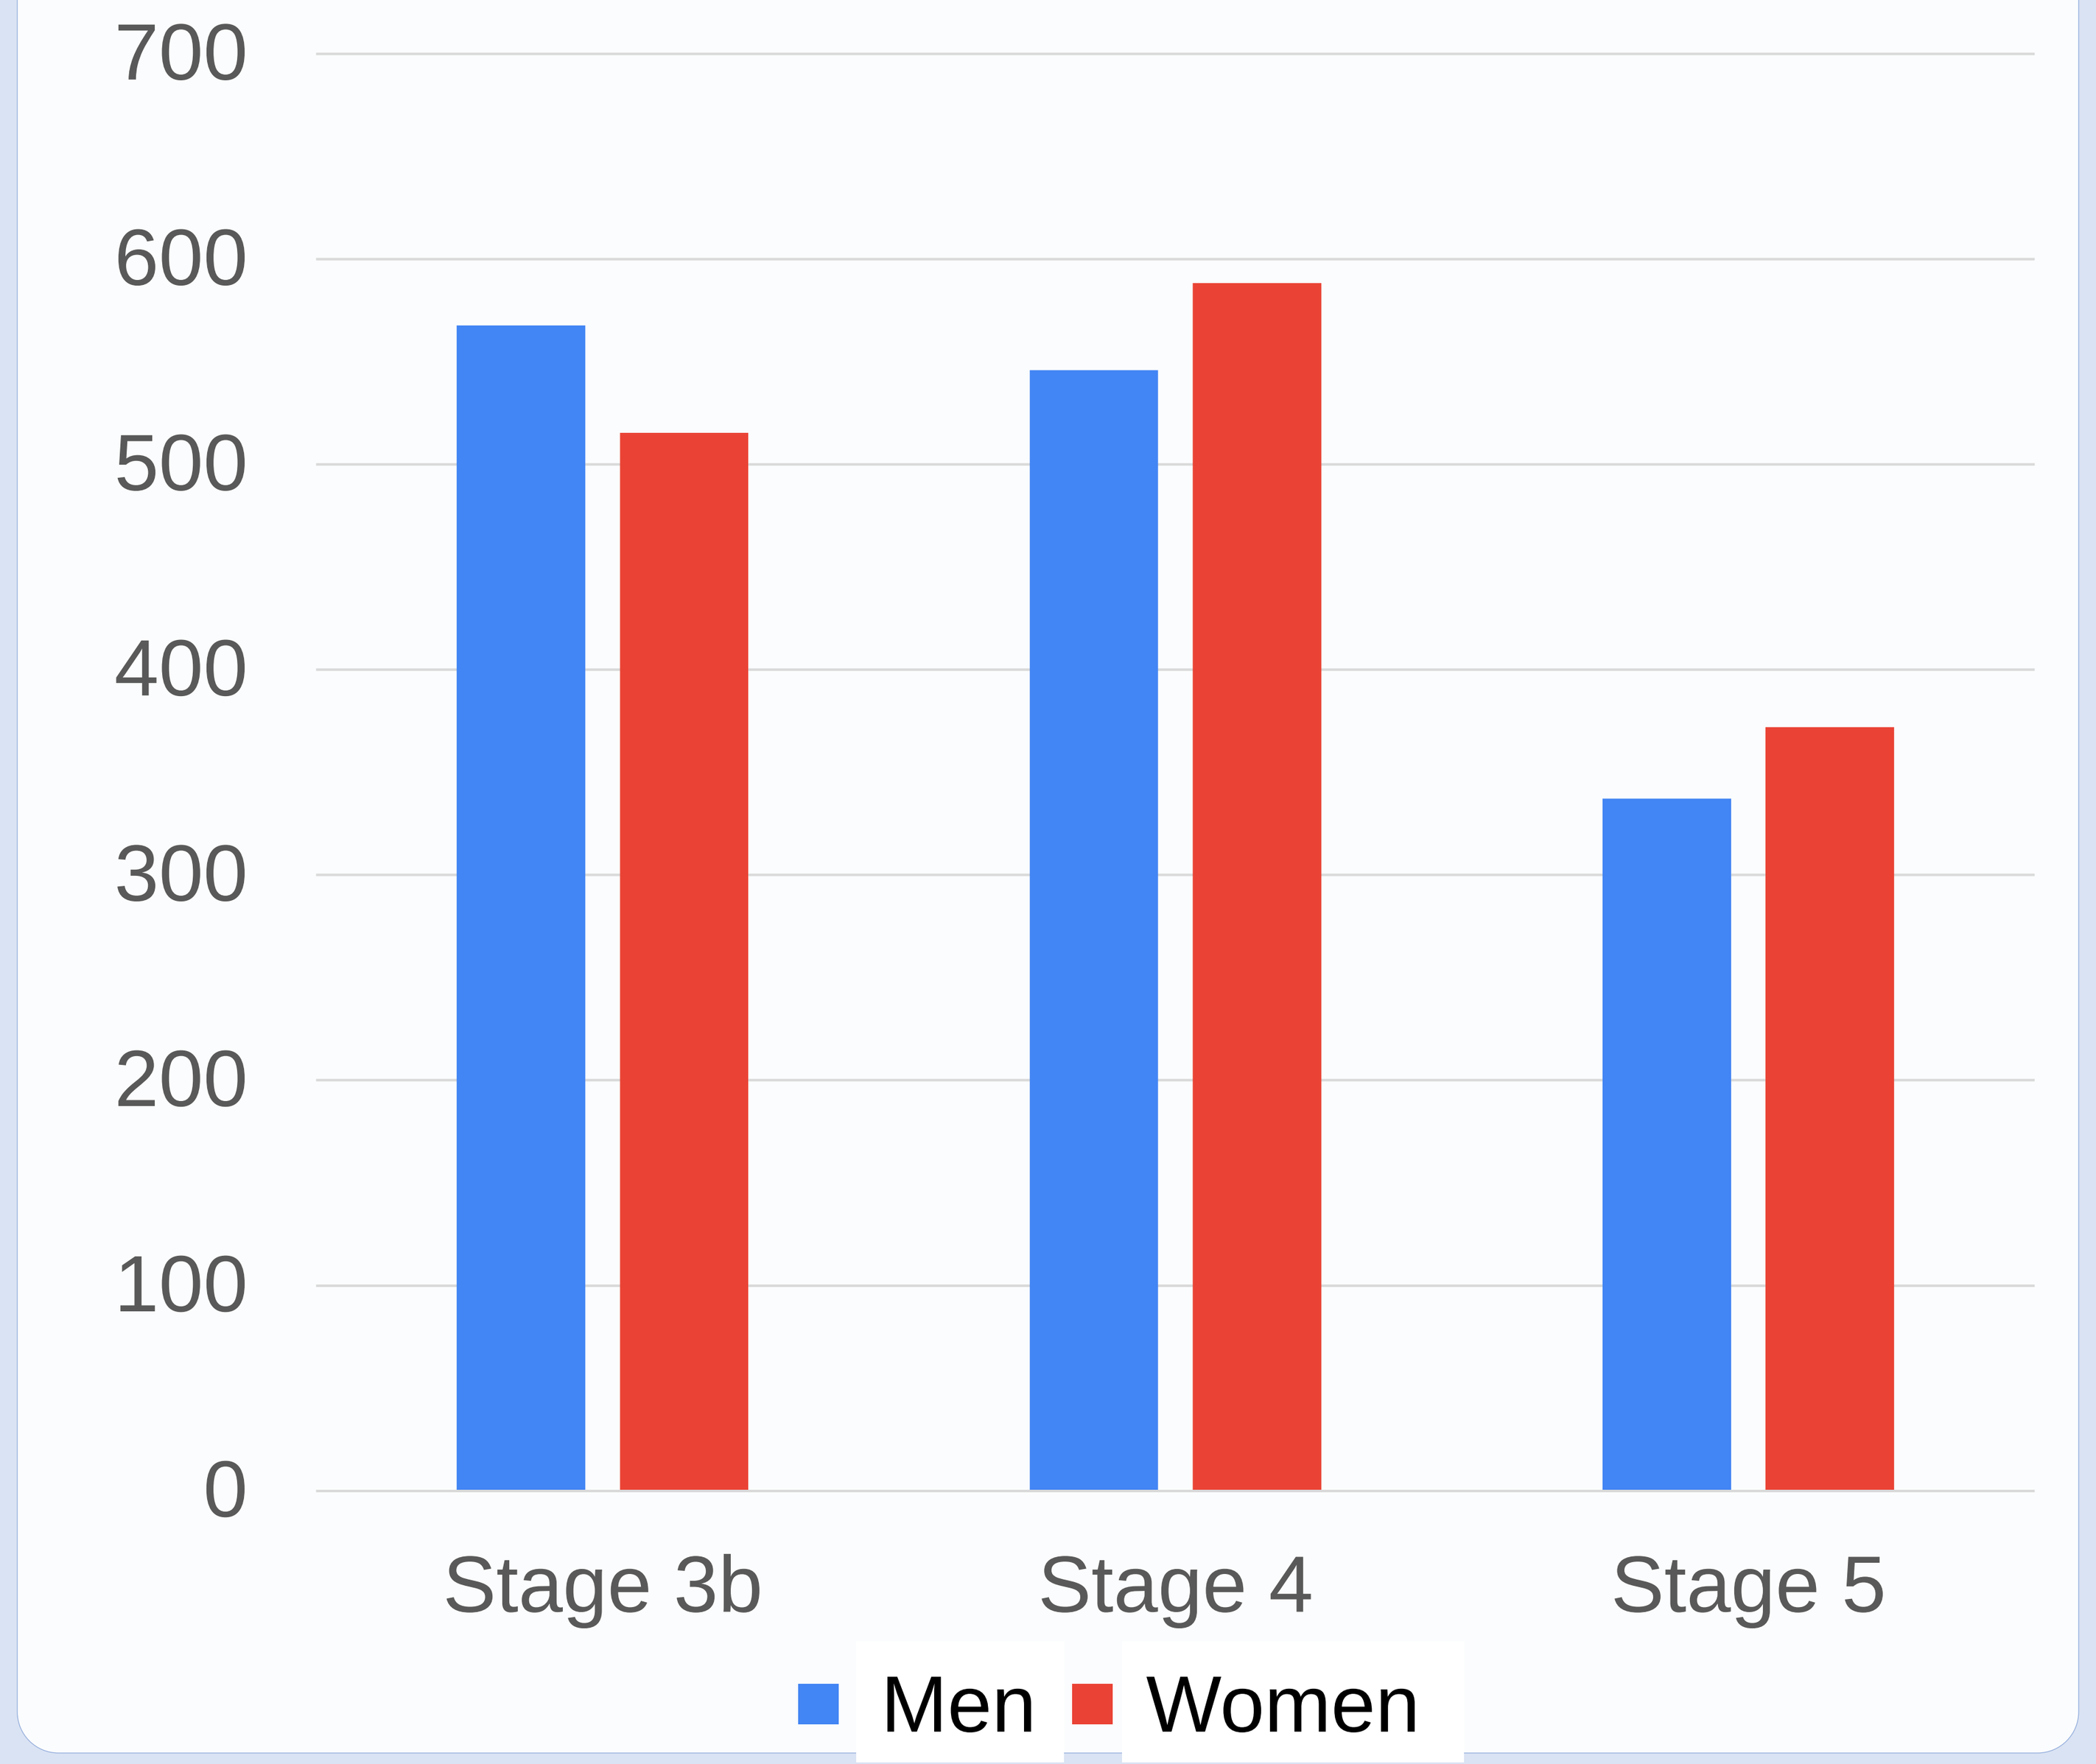
700
600
500
400
300
200
100
0
Stage 3b
Stage 4
Stage 5
Men
Women

5. Discussion

- We confirmed the higher prevalence of CKD among women. In contrast to previous studies, the difference in progression to dialysis in males was slightly faster in men than in women.
- Strengths of our study include a large sample size and rigorous application of methodology for RWD.
- Limitations of the study include the lack of information about provider and adherence to prescription of therapies to slow CKD progression (e.g., ACEi/ARBs and the SGLT2i) or the prevalence of X-linked diseases (i.e., Alport syndrome).
- Limitation of RWD include large attrition across inclusion requirements, inconsistent eGFR sampling, selection bias for resources and access to care in health systems.

6. Conclusions

Analysis using RWD confirmed: CKD was more common among women compared to men, initiation of dialysis was more common among men and they initiated more quickly. Additional research identifying biological and psychosocial factors responsible for gender differences in CKD progression and administration of CKD care is needed.

References

1. Ricardo AC, et al.: Sex-Related Disparities in CKD Progression. J AM Soc Nephrol 30: 137-146, 2019.
2. Menon V, et al.: Long-term outcomes in nondiabetic chronic kidney disease. Kidney Int 73: 1310–1315, 2008
3. FDA. U.S Department of Human and Health Services (2018). Framework for FDA's Real-World Evidence Program.

Poster Contact

Joseph Stavas, joseph.stavas@prokidney.com

RESEARCH POSTER PRESENTATION TEMPLATE © 2019
www.PosterPresentations.com